Characterization of the functional efficacy of KB-1754, a novel Kv7 activator, on smooth muscle contractility

Zena Wehbe, Elizabeth Forrester, Kelly Picchione*, Alexander Komarov*, Andrew M Inglis*, Lynn Resnick*, James Hale*, Michael Bozik*, Steven Dworetzky*, Iain A Greenwood

Institute of Molecular and Clinical Sciences, St. George's University of London, UK
*Biohaven Pharmaceuticals, New Haven, CT, USA

Introduction

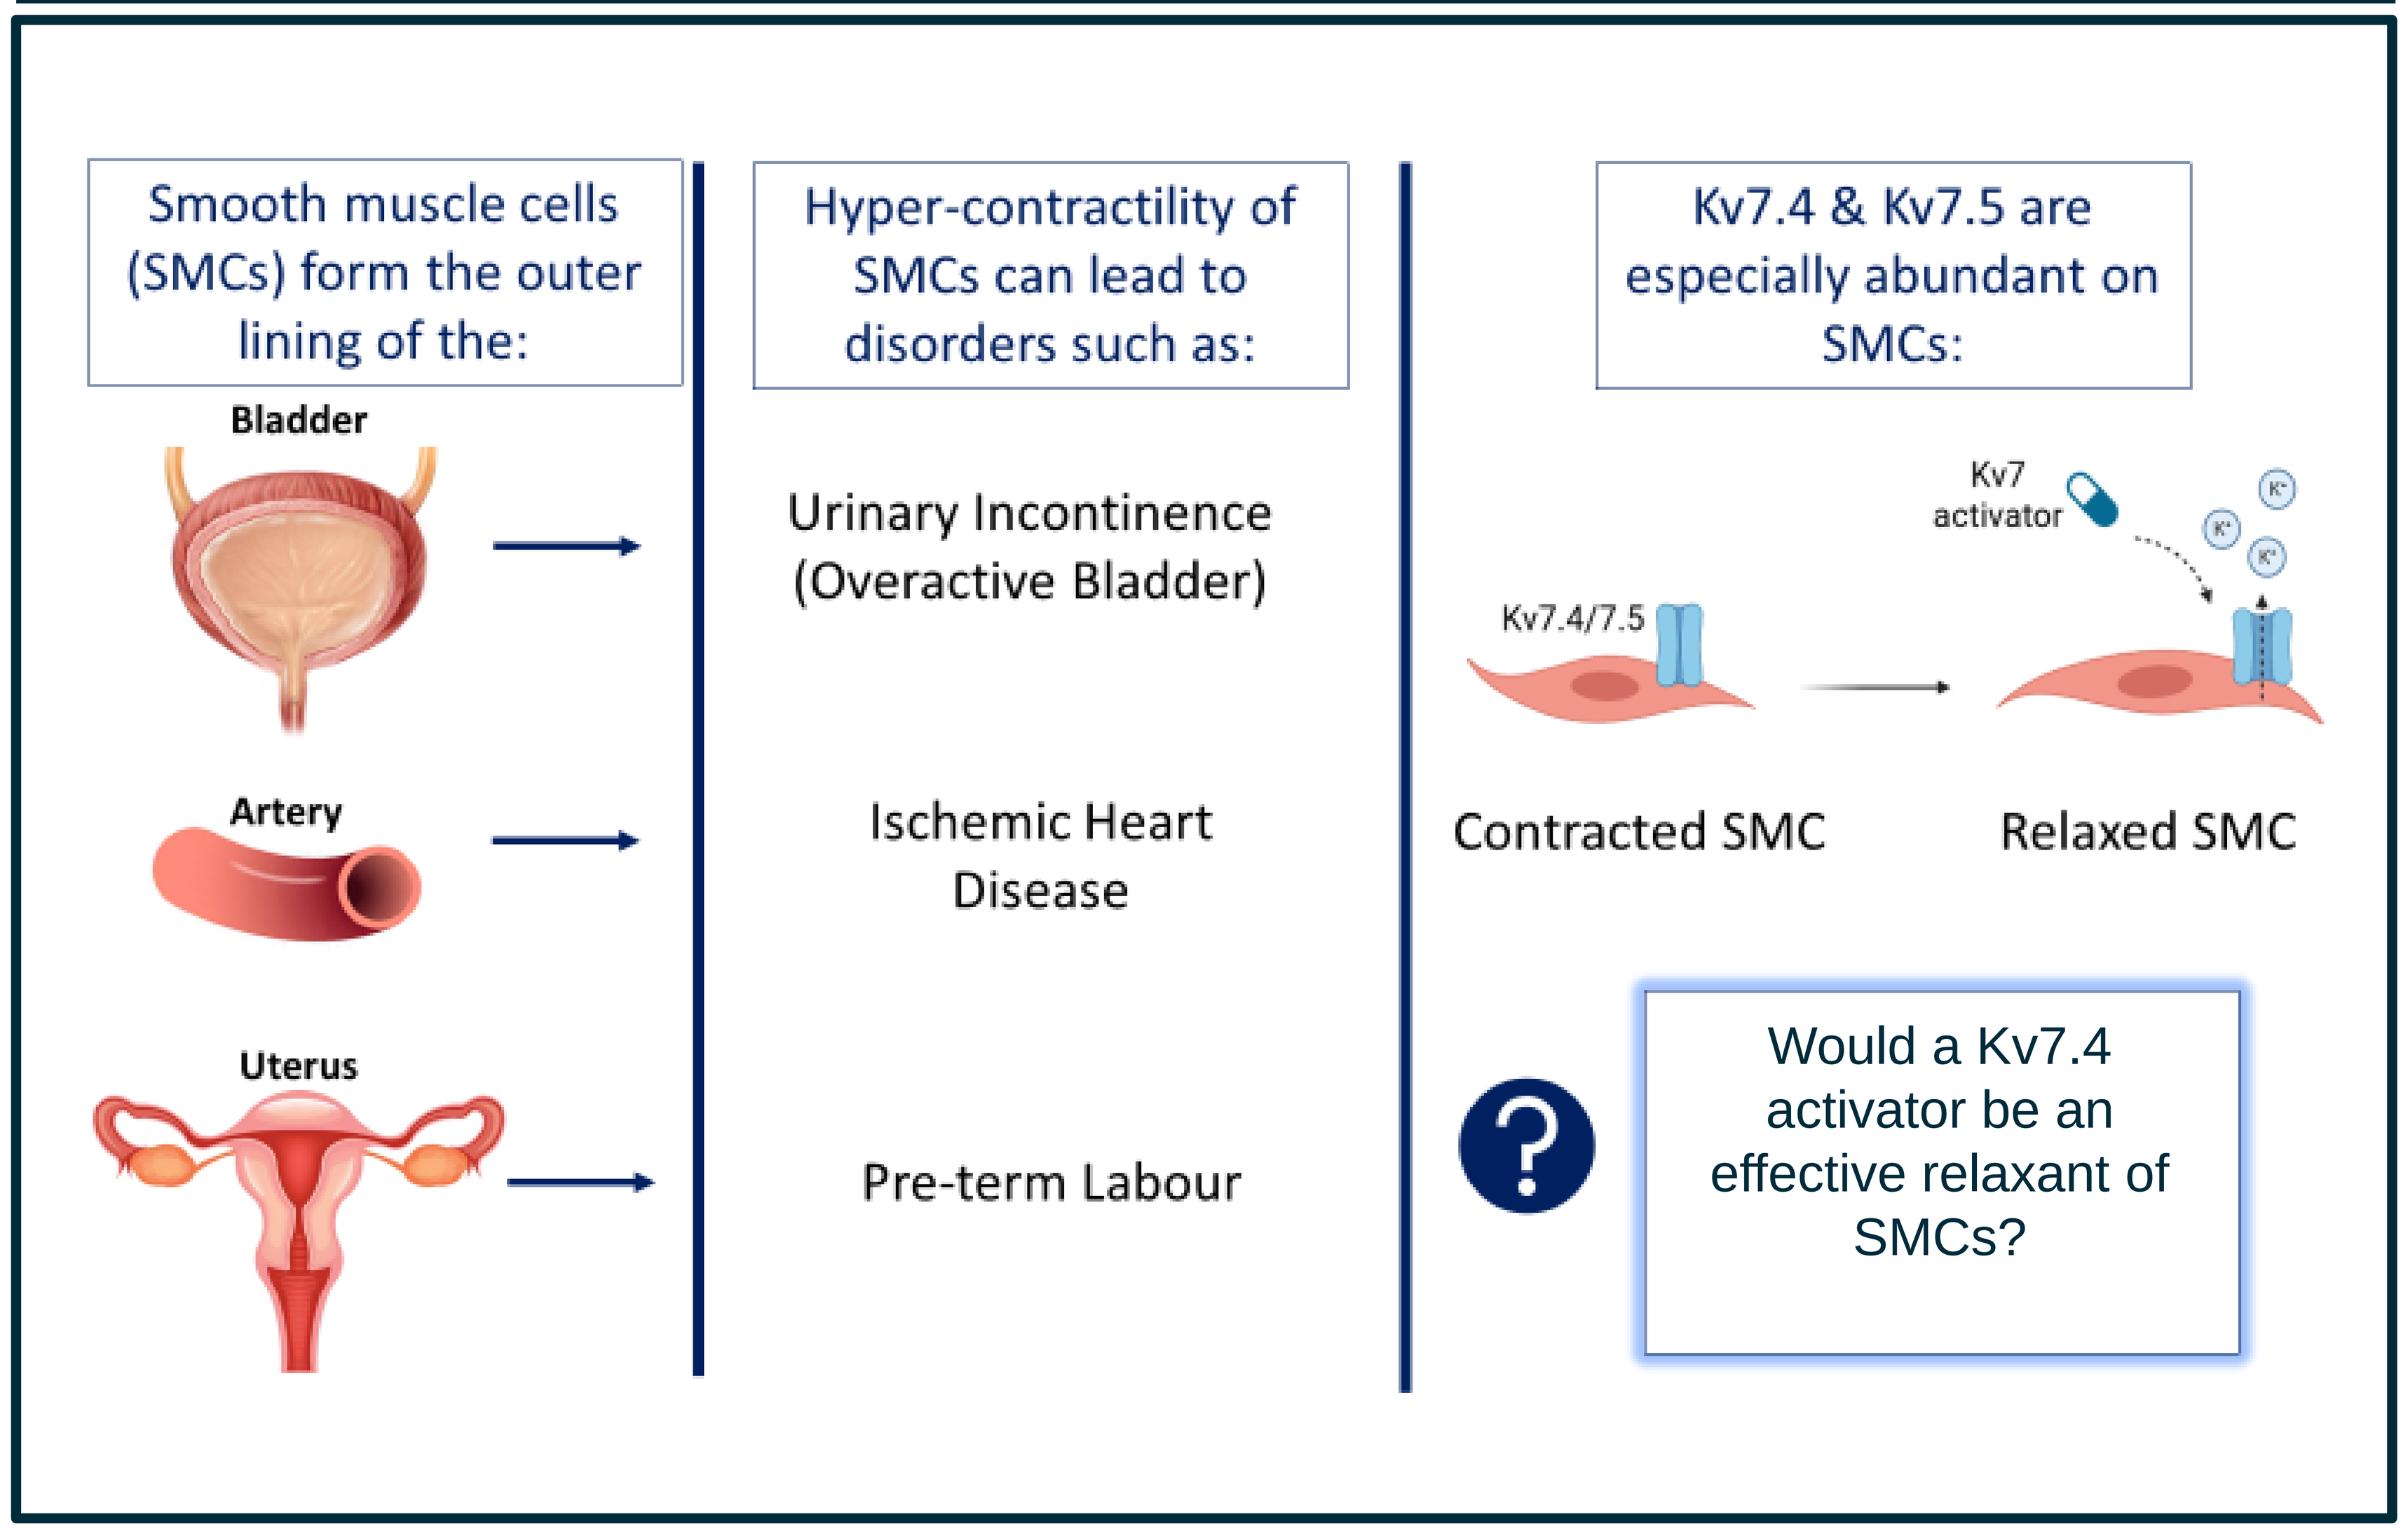
Smooth muscle cells (SMCs) form the outer lining of the:
Bladder
Artery
Uterus
Hyper-contractility of SMCs can lead to disorders such as:
Urinary Incontinence (Overactive Bladder)
Ischemic Heart Disease
Pre-term Labour
Kv7.4 & Kv7.5 are especially abundant on SMCs:
Kv7 activator
Kv7.4/7.5
Contracted SMC
Relaxed SMC
Would a Kv7.4 activator be an effective relaxant of SMCs?

Gap

Selective Kv7.4 activators have not been extensively reported or studied in SMCs. Given the differential abundance of this channel on SMCs, it is unknown if activation of Kv7.4 would be effective in counteracting various conditions of smooth muscle hypercontractility.

Hypothesis

We hypothesize that a Kv7.4 activator will be an effective relaxant in various smooth muscle tissues, like the bladder, mesenteric arteries and uterus.

Aims

The present study aimed to characterize the functional efficacy of a Kv7.4 activator, KB-1754, in different tissues and under various conditions like concentration, time and sex. In addition, we compared its efficacy to retigabine.

Methods

Whole-cell, voltage-clamp experiments were performed on the Qube®, an automated patch-clamp system. Compounds were tested at 10 µM. Peak inward tail currents were measured at 0 mV after activating pulses (-110 to +30 mV). Mean data were fit with a Boltzmann sigmoid equation to calculate the half-maximal voltage (Vhalf) and max tail current. Strips of bladder from male and female Wistar rats were mounted in a 4 chamber myograph to measure and display contractile amplitudes of the tissues. Functional studies of KB-1754 were conducted by the addition of the drug to the tissue chambers of the myograph. Changes to contractile amplitude (in milli-Newtons) were measured and plotted using GraphPad Prism software. Expression studies were conducted using reverse transcription quantitative polymerase chain reaction (RT-qPCR).

Spontaneous & nerve-evoked (electric field stimulated or EFS) contractions

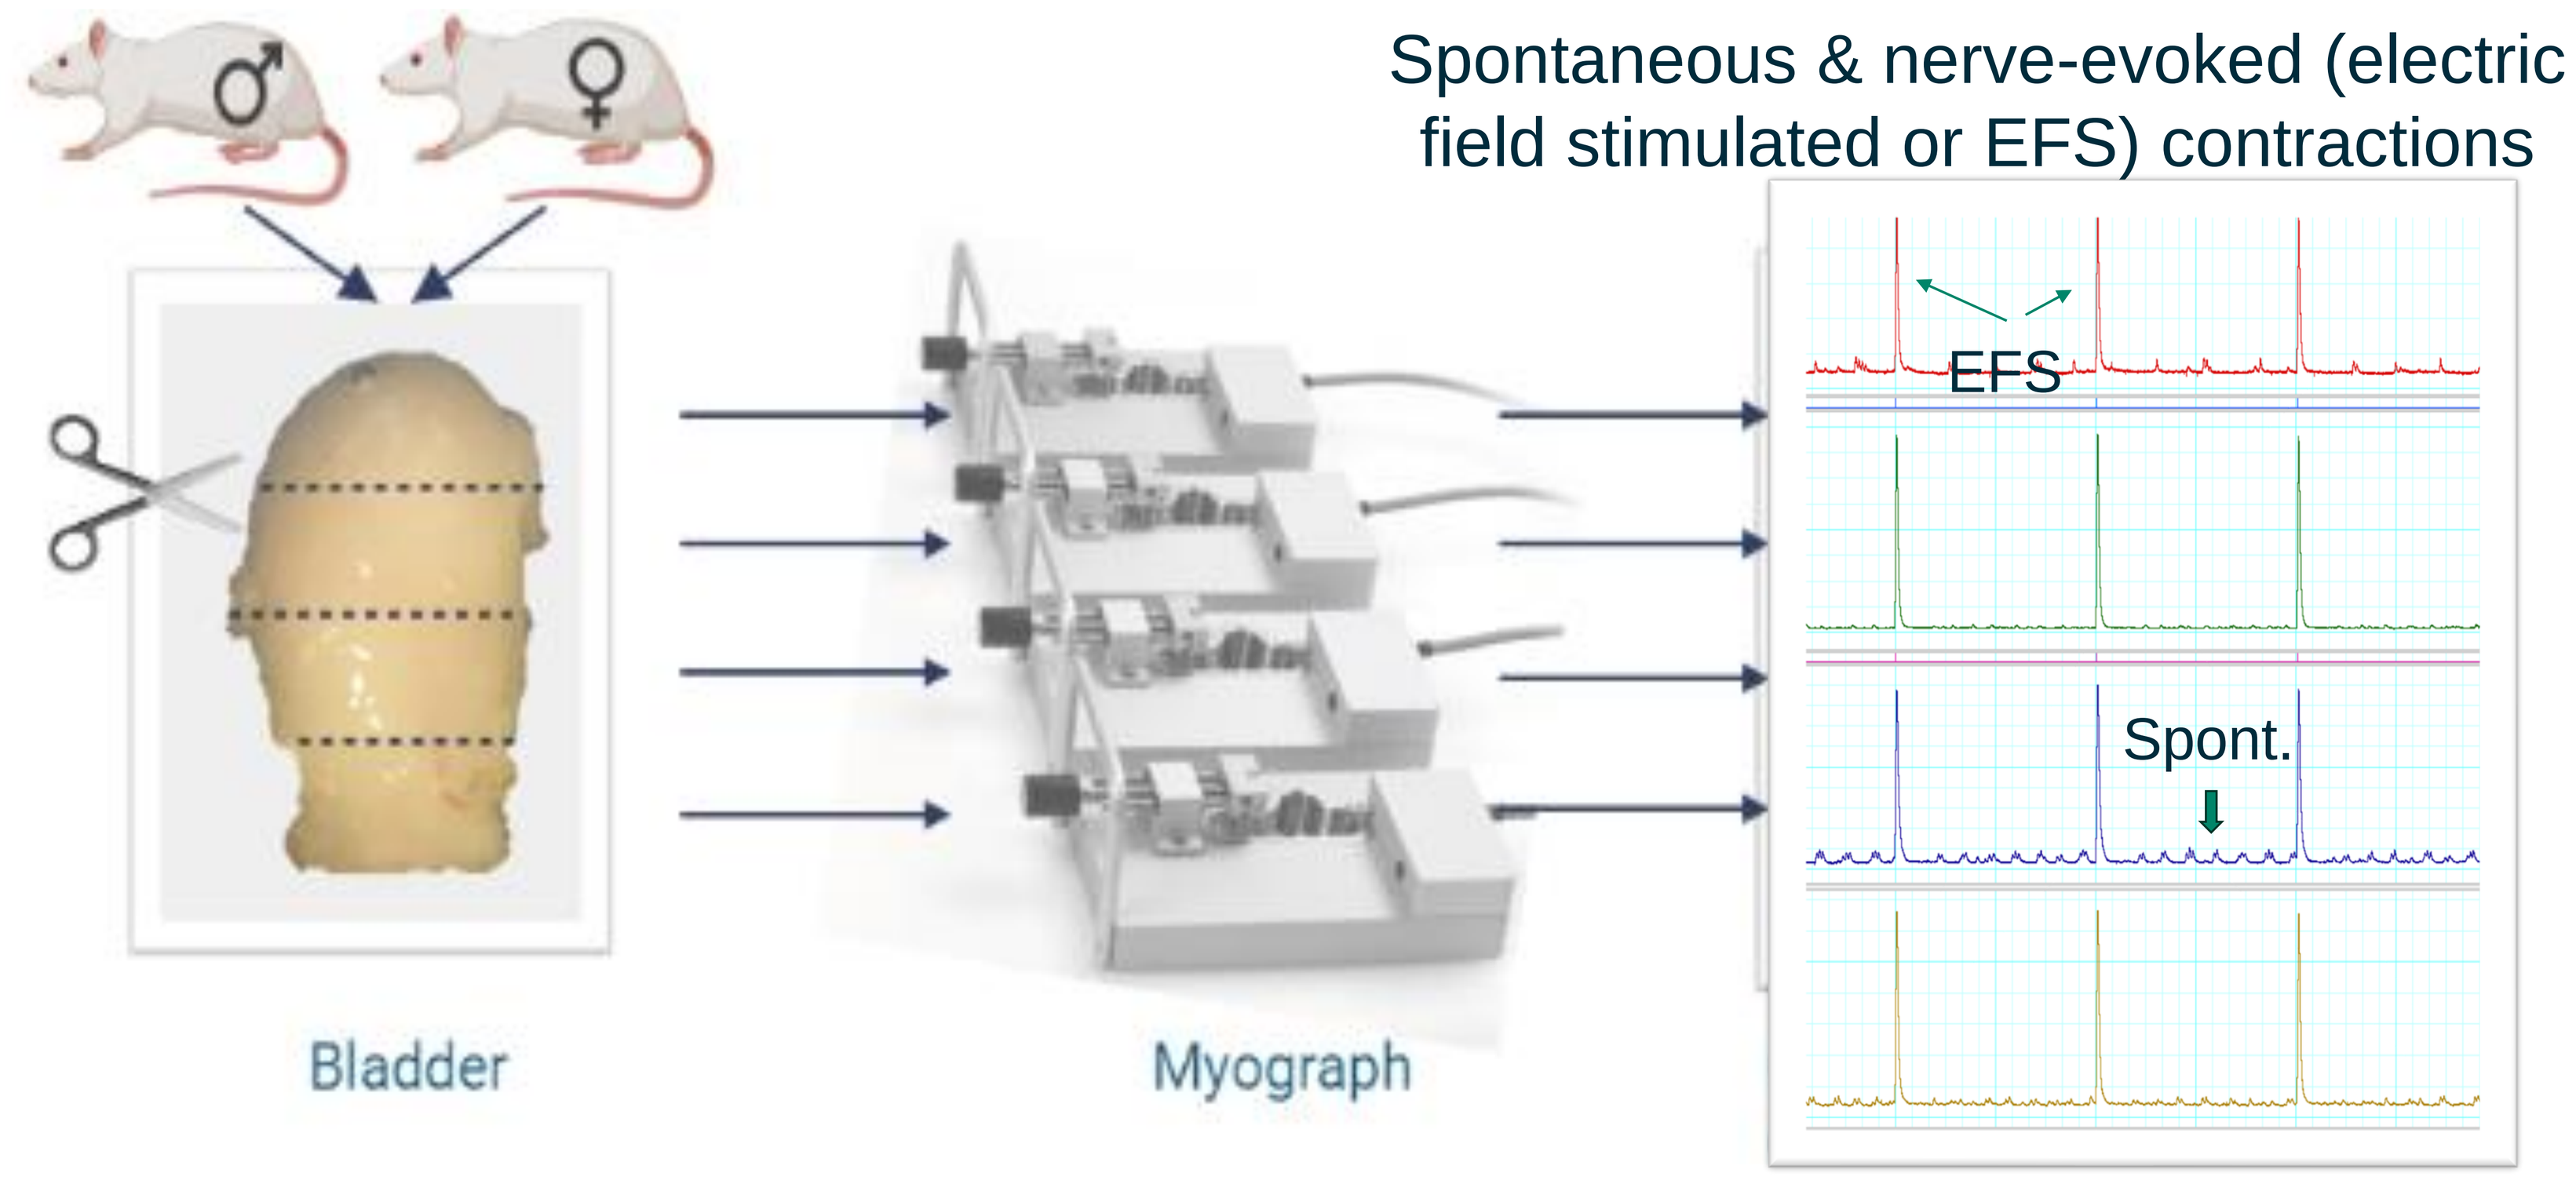
EFS
Spont.
Bladder
Myograph

Conclusion

KB-1754 preferentially activates Kv7.4 when tested in CHO cells stably expressing Kv7.4 or Kv7.2/7.3. KB-1754 relaxed the detrusor, uterus and mesenteric arteries in a concentration-dependent manner and exerted significantly greater relaxation in bladders from males compared to proestrous females. The effects of KB-1754 were maintained in conditions of hypercontractility in both the detrusor and uterus. KB-1754 is an effective Kv7.4 prototype activator to better understand the role of this channel in smooth muscle contractility.

Results

1. KB-1754 preferentially activates Kv7.4 as shown by electrophysiological studies.

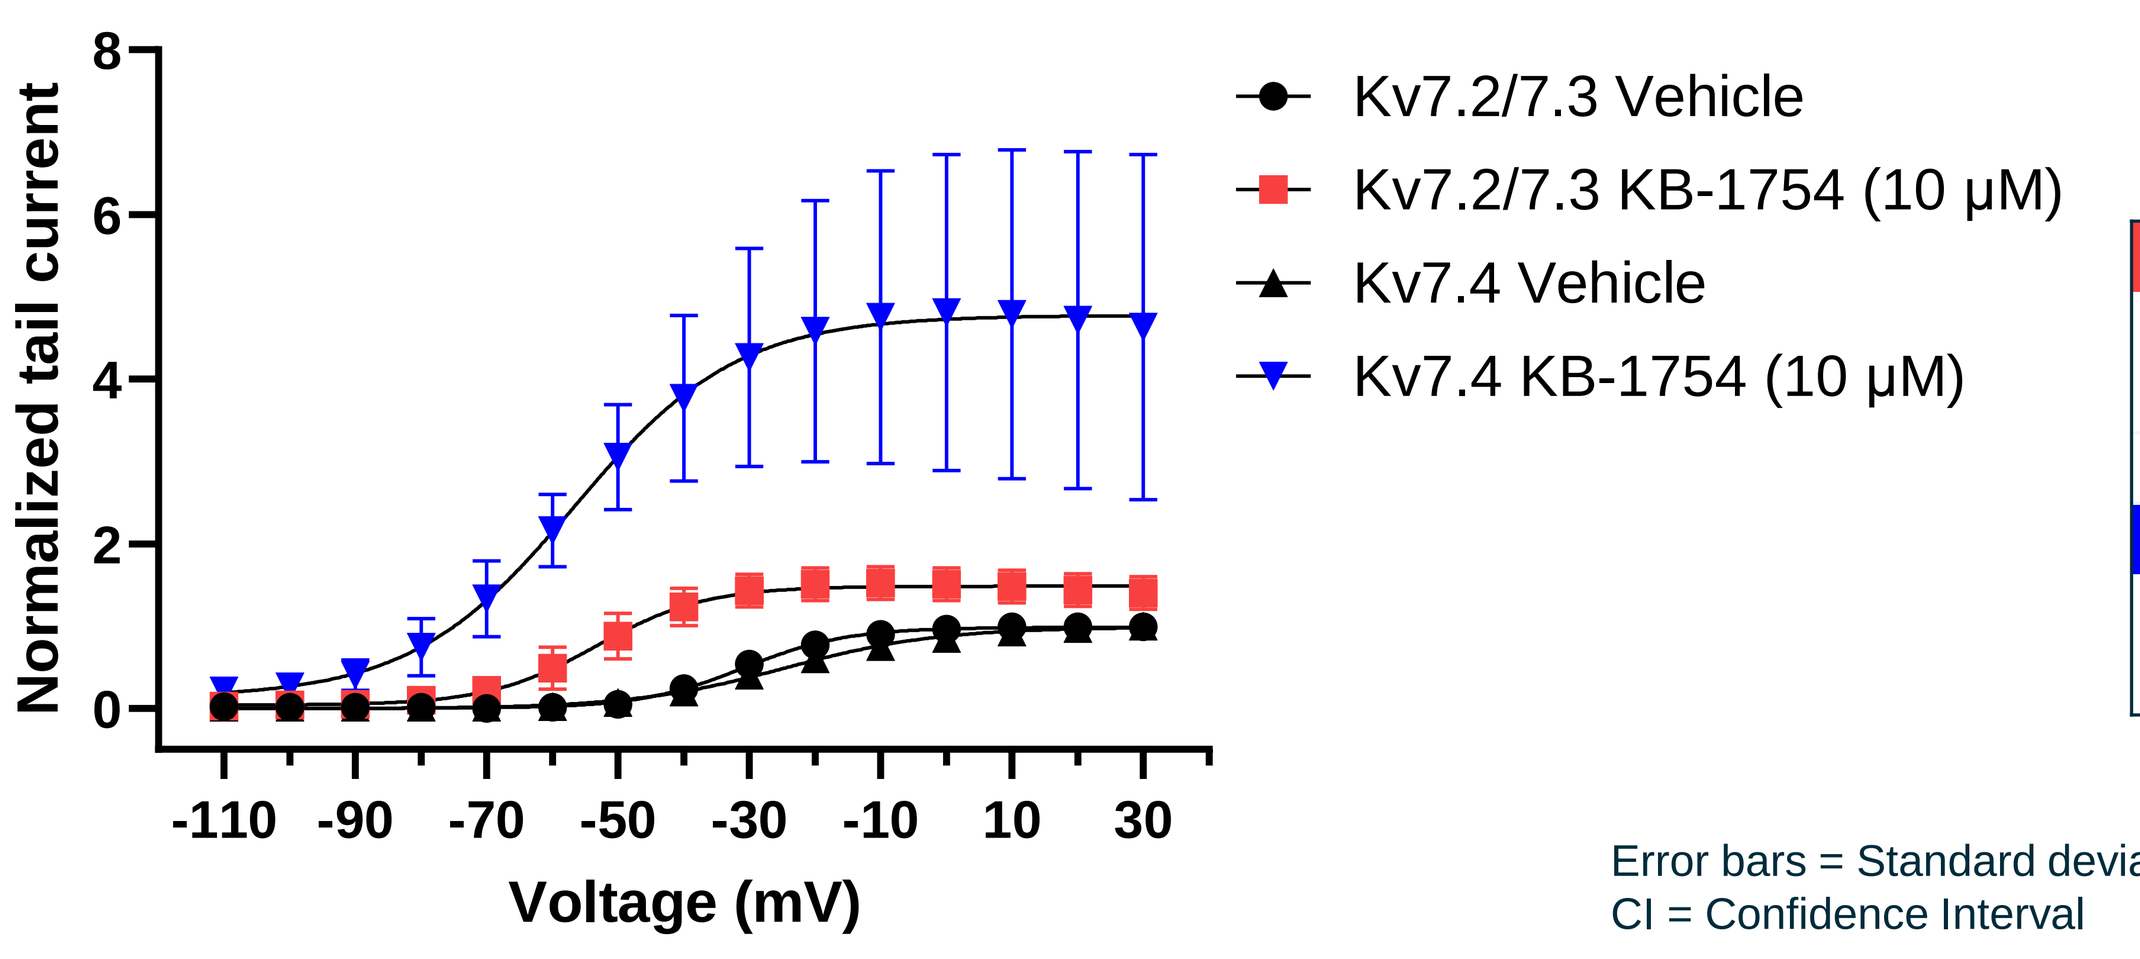
Normalized tail current
Voltage (mV)
Kv7.2/7.3 Vehicle
Kv7.2/7.3 KB-1754 (10 µM)
Kv7.4 Vehicle
Kv7.4 KB-1754 (10 µM)

Delta Vhalf = Vhalf compound – Vhalf vehicle
Normalized tail = Tail compound – Tail vehicle

Kv7.2/7.3	n	Delta Vhalf (mV), 95% CI	Normalized Tail, 95% CI
KB-1754	24	-22 (-24 to -21)	1.5 (1.5 to 1.5)
Retigabine	108	-33 (-34 to -32)	1.1 (1.1 to 1.2)

Kv7.4	n	Delta Vhalf (mV), 95% CI	Normalized Tail, 95% CI
KB-1754	20	-32 (-37 to -27)	4.8 (4.5 to 5.1)
Retigabine	109	-9 (-10 to -7)	2.8 (2.7 to 2.8)

Error bars = Standard deviations
CI = Confidence Interval

2. The effect of KB-1754 on rat bladder is more potent on spontaneous versus nerve-evoked (EFS) contractions (A). KB-1754 is more potent than the Kv7 activator, retigabine, on both spontaneous and EFS contractions (B,C).

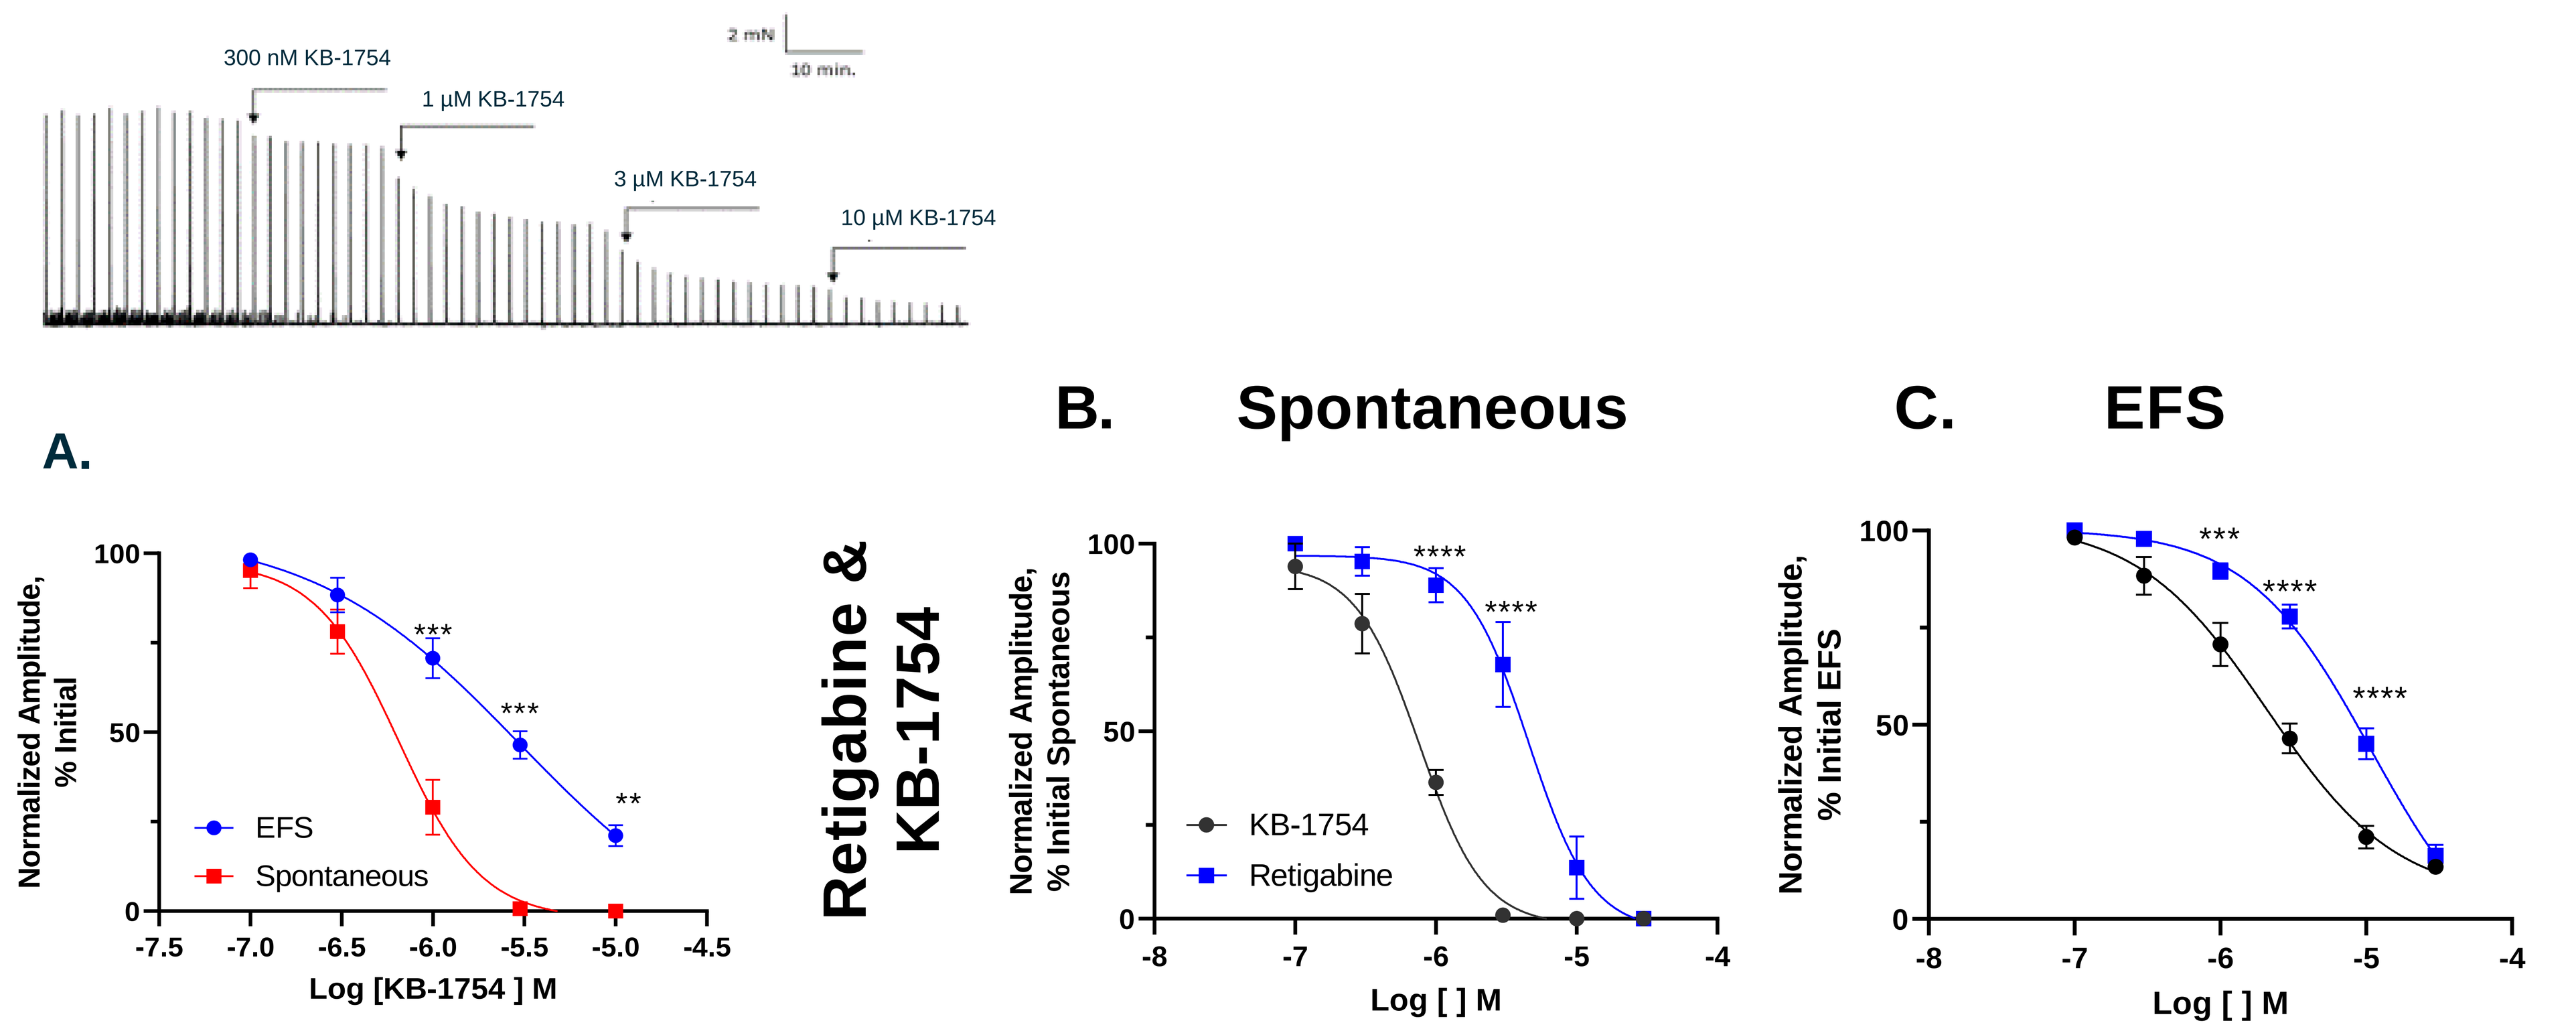
300 nM KB-1754
1 µM KB-1754
3 µM KB-1754
10 µM KB-1754
2 mN
10 min.
A.
Normalized Amplitude, % Initial
EFS
Spontaneous
Log [KB-1754] M
B. Spontaneous
C. EFS
Retigabine & KB-1754
Normalized Amplitude, % Initial Spontaneous
KB-1754
Retigabine
Log [] M
Normalized Amplitude, % Initial EFS

N=5, Two-Way ANOVA, SEM, Bonferroni ad hoc, *p<0.05, **p<0.01,***p<0.0001,****p<0.0001

3. KB-1754 is effective in hypercontractile conditions, including those induced by capsaicin (A) and ERG channel blockade E4031 (B). KB-1754 was least potent in bladders from proestrous and oestrous females (PE) compared to males and D.M females (C).

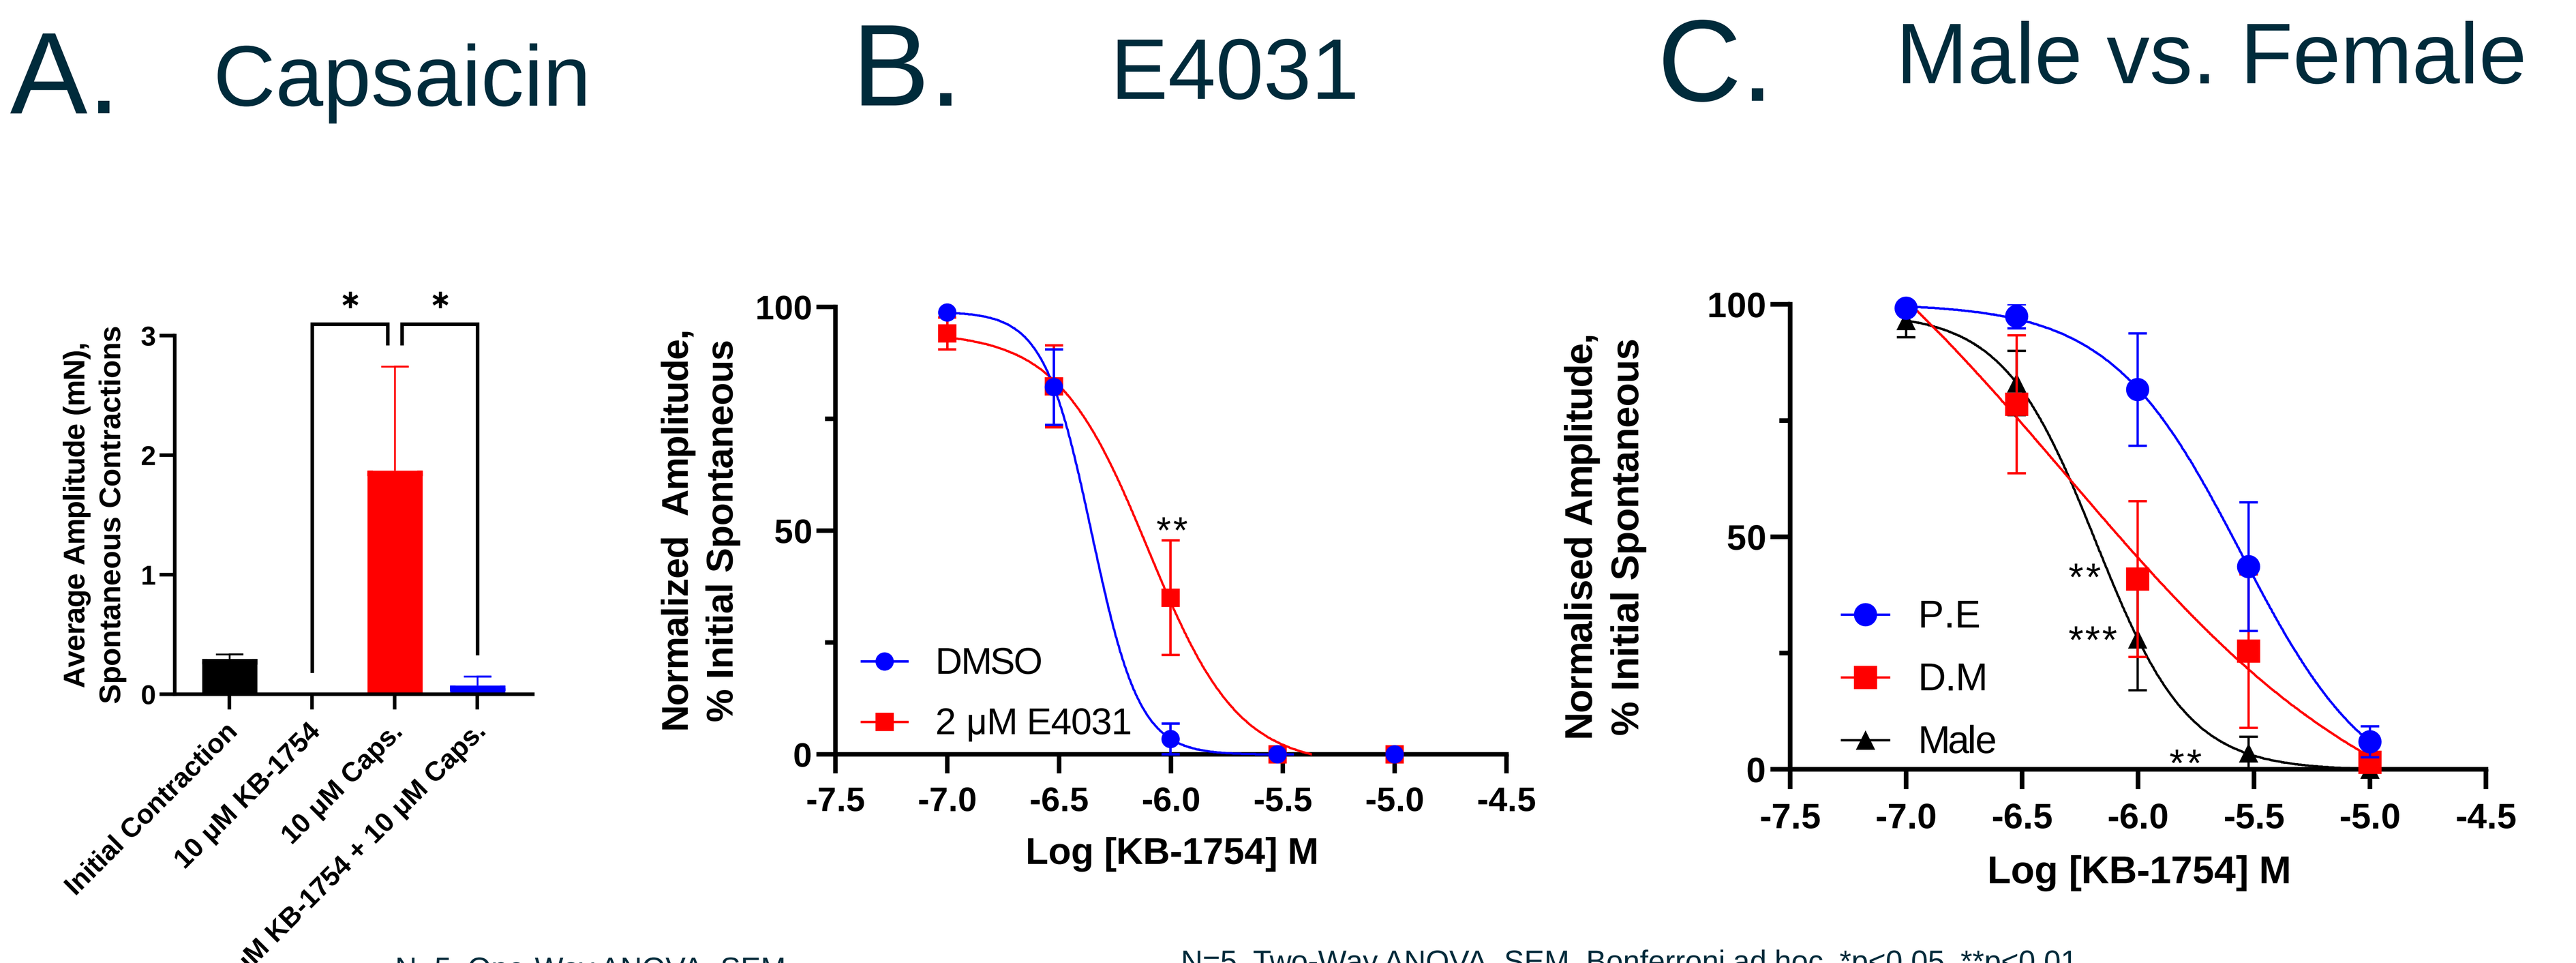
A. Capsaicin
B. E4031
C. Male vs. Female
Average Amplitude (mN), Spontaneous Contractions
Initial Contraction
10 µM KB-1754
10 µM Caps.
10 µM KB-1754 + 10 µM Caps.
Normalized Amplitude, % Initial Spontaneous
DMSO
2 µM E4031
Log [KB-1754] M
Normalised Amplitude, % Initial Spontaneous
P.E
D.M
Male

N=5, One-Way ANOVA, SEM

N=5, Two-Way ANOVA, SEM, Bonferroni ad hoc, *p<0.05, **p<0.01

4. KB-1754 is also an effective relaxant of the uterus (A) and mesenteric arteries (B).

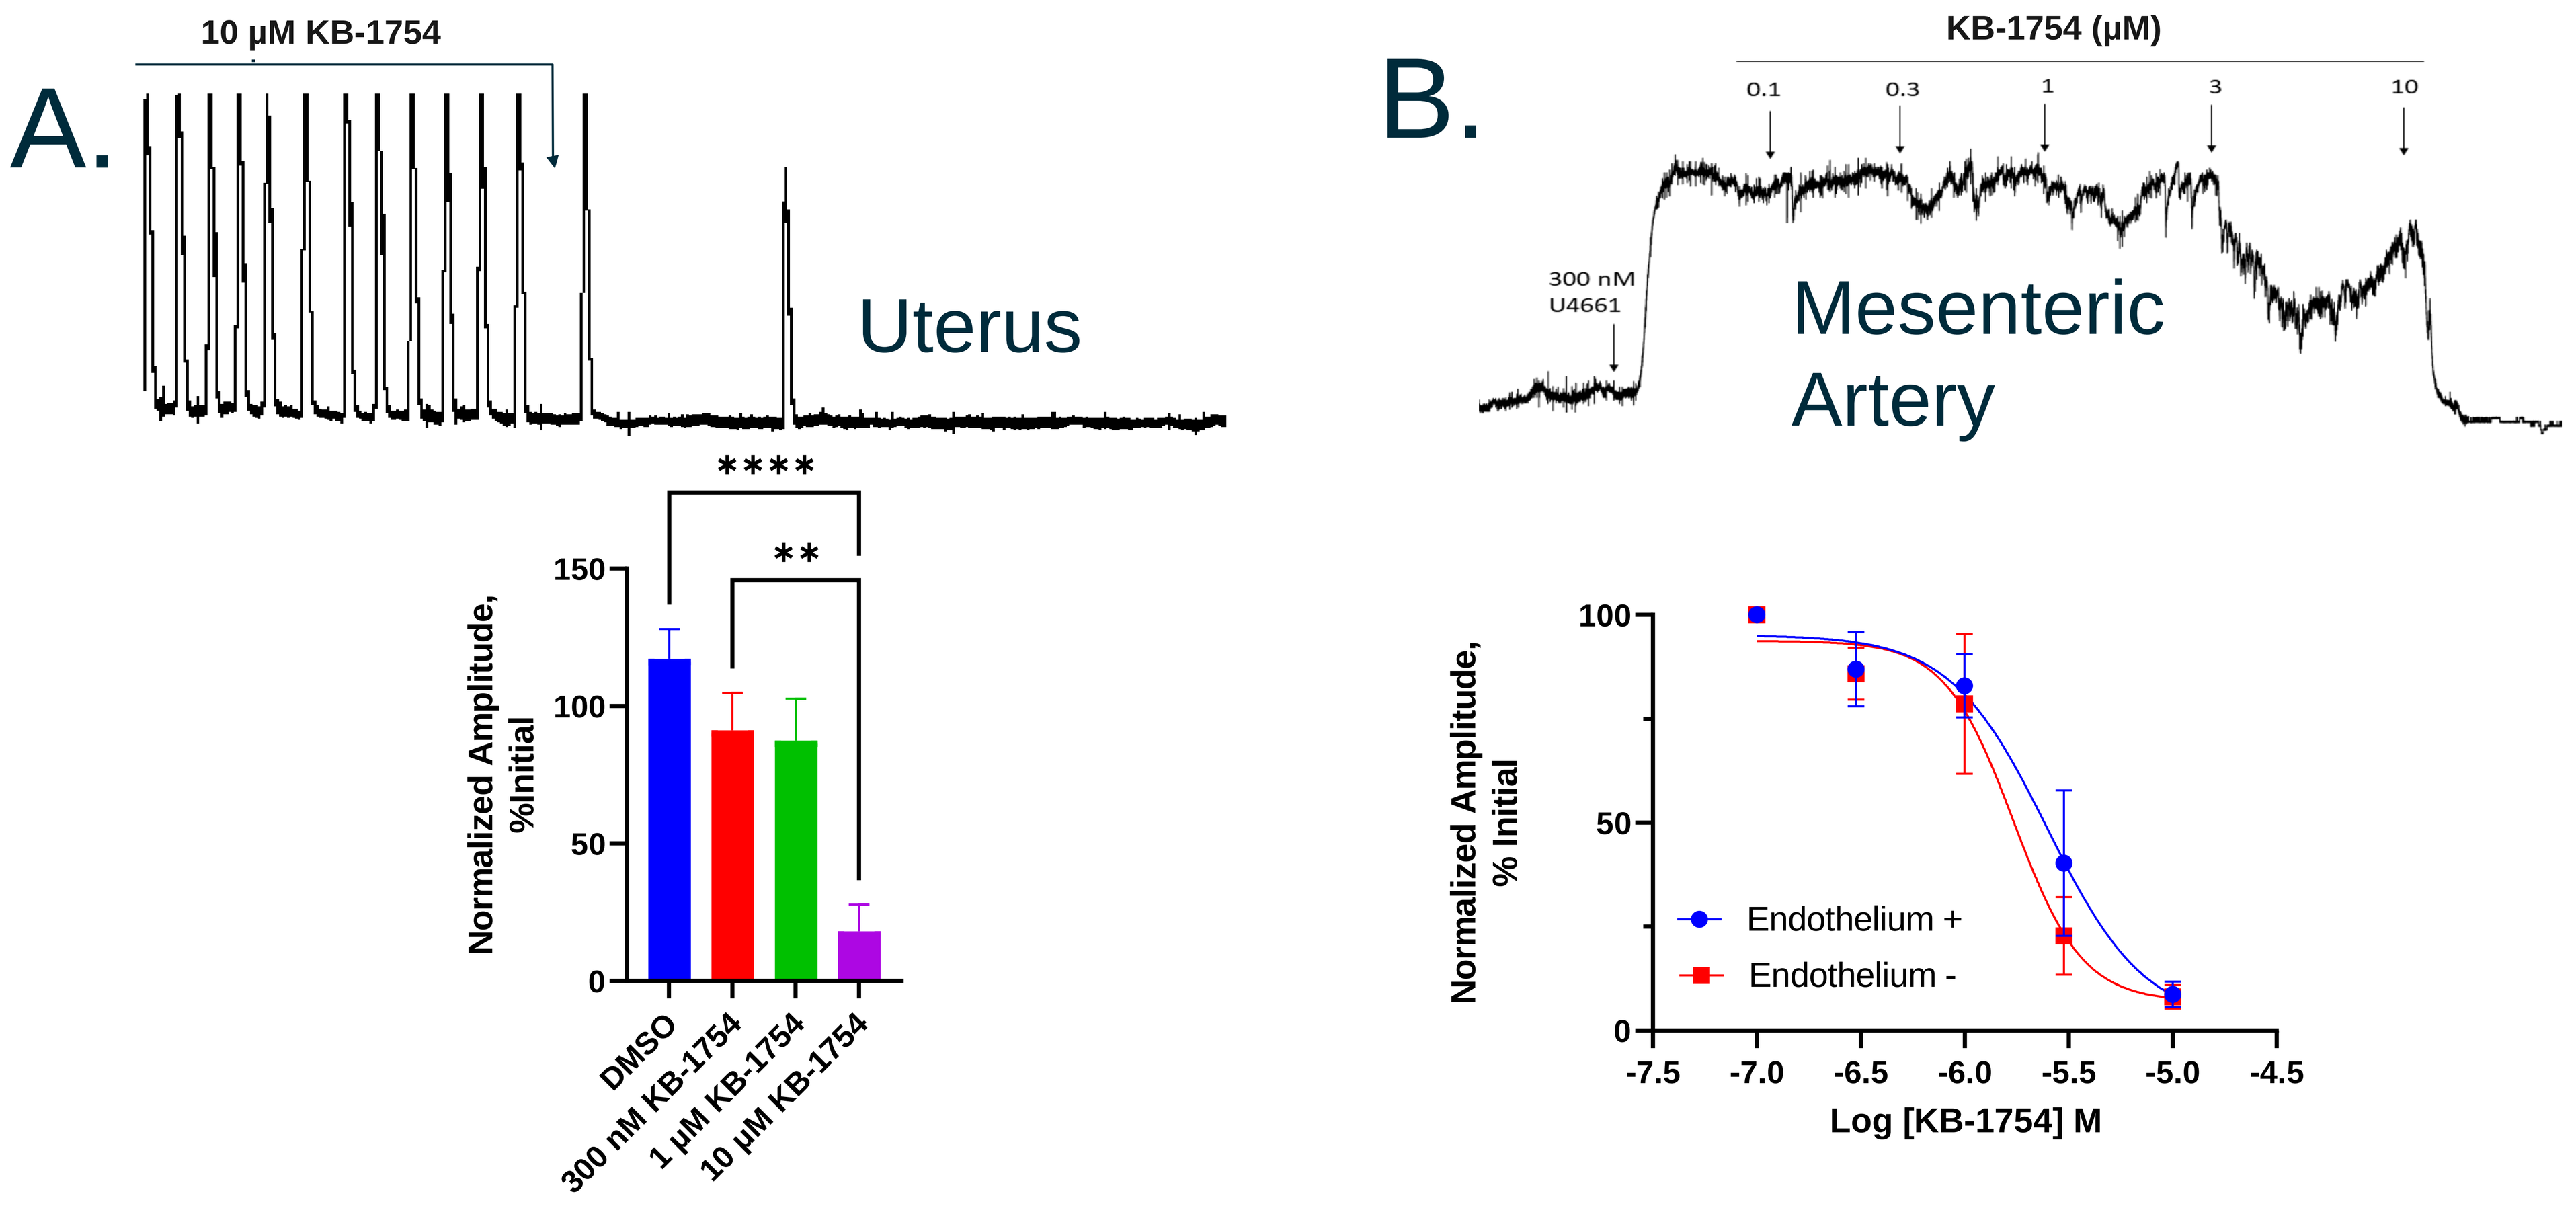
A.
10 µM KB-1754
Uterus
Normalized Amplitude, %Initial
DMSO
300 nM KB-1754
1 µM KB-1754
10 µM KB-1754
B.
KB-1754 (µM)
0.1
0.3
1
3
10
300 nM U4661
Mesenteric Artery
Normalized Amplitude, % Initial
Endothelium +
Endothelium -
Log [KB-1754] M

N=5, One-Way ANOVA(left), unpaired t test (right), SEM, **p<0.01, ****p<0.0001

N=5, Two-Way ANOVA, SEM

5. The effect of KB-1754 is inhibited in the presence of non-selective Kv7 inhibitors, Linopirdine and XE991.

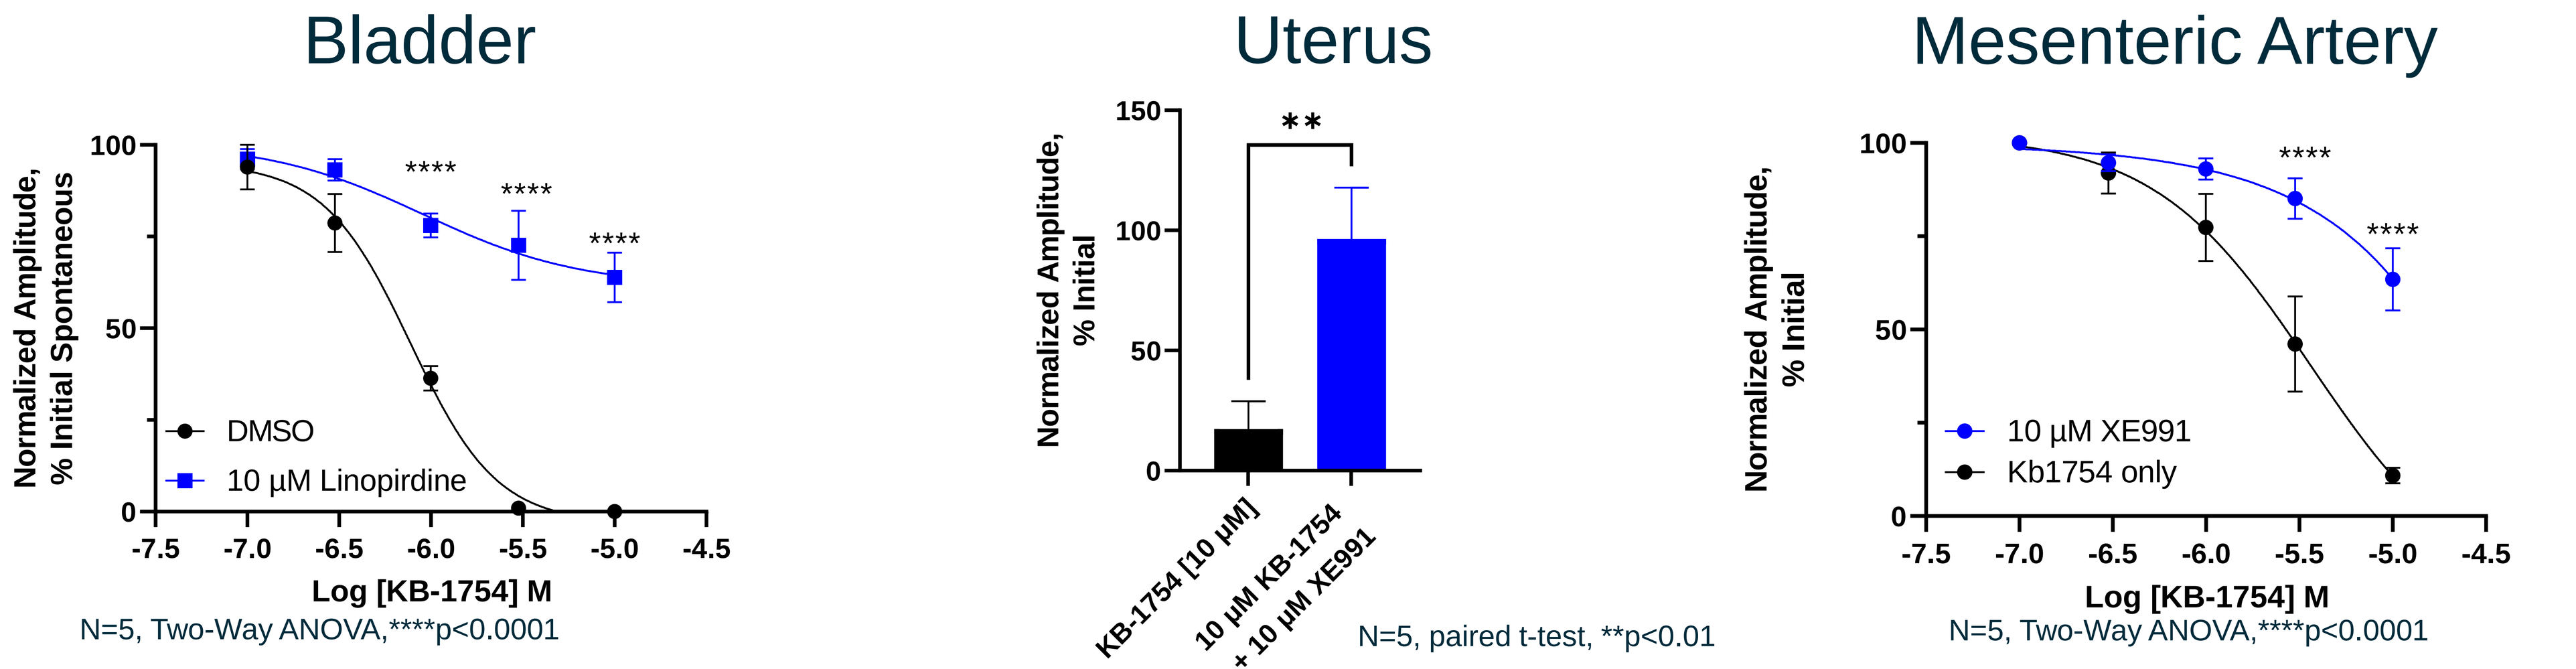
Bladder
Uterus
Mesenteric Artery
Normalized Amplitude, % Initial Spontaneous
DMSO
10 µM Linopirdine
Log [KB-1754] M
Normalized Amplitude, % Initial
KB-1754 [10 µM]
10 µM KB-1754 + 10 µM XE991
10 µM XE991
Kb1754 only

N=5, Two-Way ANOVA,****p<0.0001

N=5, paired t-test, **p<0.01

N=5, Two-Way ANOVA,****p<0.0001